Comparison of Homogenization Techniques for the Extraction of Total RNA from Mammalian Tissues

ROSETTA INPHARMATICS

Utilizing Focused Acoustic Energy for Tissue Disruption – A Feasibility Study

Mark D. Morris, Michael R. Meyer, George Y. Tokiwa, Thomas L. Fare - Rosetta Inpharmatics LLC, a wholly owned subsidiary of Merck & Co., Inc.
401 Terry Avenue N Seattle, WA 98109

Abstract

Here we describe a process that is capable of extracting RNA from a broad range of tissue types and masses that utilizes focused acoustic energy to homogenize frozen tissue. Employing the acoustic tissue disruption process, non-contact homogenization can be performed through a computer controlled method, significantly increasing the efficiency of the homogenization, while at the same time allowing for an increased throughput capacity of samples. We compare this non-contact tissue disruption process, using the Covaris E200 instrument, with the more commonly used rotor stator (Polytron) tissue disruption technology. Total RNA sample integrity and RNA yield from the two processes are compared, as well as reproducibility of the homogenization. Input of the isolated RNA into sensitive downstream applications, such as microarrays, will also be addressed.

Introduction

Tissue homogenization is a bottleneck for high throughput sample processing
High throughput experimentation in genomics, proteomics, and metabolic profiling requires high throughput and reproducible sample preparation methods. In particular, existing tissue homogenization methods such as the rotor stator (Polytron) method are labor intensive, not automation amenable and have limited scalability. In addition, this method is susceptible to cross contamination of samples due to carry-over from sample to sample. This problem would be especially detrimental for sensitive downstream applications such as quantative PCR or microarray applications.

The common technologies for tissue homogenization currently in use include the mortar and pestle, bead mill, or the previously mentioned rotor stator. The mortar and pestle is the traditional favorite due to its effectiveness across a broad range of tissue types. However, the mortar and pestle is extremely labor intensive making it difficult to scale up for high throughput. It can also be employed as the first step in combination with the Polytron (Brinkmann). The bead mill is available in a multi-well format and is effectively high throughput, but it does have limitations for volume and mass. The multi-well bead mill is also restricted to use the same homogenization program for each plate every well in the plate limiting the tissue type(s) that can be processed at the same time. The Polytron device uses an outer fixed shaft, or stator, with an inner rotor. The sample is vigorously mixed and pressed through the narrow slots between the rotor and stator, thus having a shearing effect . The shearing is effective for homogenization of tissues, but the probe is very difficult to clean between subsequent samples and thus can be a source of cross contamination. Polytron type devices have limited control of the conditions under which homogenization occurs, including controlling temperature, the time of homogenization and revolutions of the rotor, all of which are often imprecise.

Focused Acoustic Energy Tissue Disruption Technology
As an alternative to the other mentioned tissue homogenization methods, Covaris Inc. (Woburn, Mass) has developed a new application for existing acoustic technology by applying it to tissue disruption. Their E200 instrument uses a transducer, which focuses sound waves through vessel walls to effectively disrupt and homogenize tissue utilizing cavitation events, the rapid creation and collapsing of bubbles on the sample surface. Scalable energy input allows for a variety of effects to be conveyed to the sample that can range from gentle mixing to the disruptive force of homogenization. The cavitation bubble that is formed to create the shockwave can also be scaled to further adjust the disruptive effect on the tissue that is being homogenized. For example, specific tissue types that are extremely fibrous may require a higher level of "bubble" enlargement for a more vigorous mixing or homogenization effect. The major benefits of this kind of technology are that the energy is computer controlled, reproducible, scalable, and the acoustics convey the disruptive effect without any apparatus physically contacting the sample. Additionally, the system is closed preventing cross contamination or the release of aerosols from the homogenate.

Comparison of Acoustic to Rotor Stator Homogenization
Here we compare the commonly applied Polytron rotor stator technology (benchmark by which to compare) with the focused energy acoustic Covaris homogenization technology for their ability to homogenize tissues for the purpose of extracting Total RNA for use with microarrays. Table 1 shows a comparison between the main features of each method and highlights the relevant issues. Here we report that the acoustic technology is comparable in its ability to reproducibly homogenize liver, muscle and brain tissue samples and that it allows for the extraction of Total RNA that is comparable in yield and quality to polytron processed samples. The Covaris acoustic technology also has advantages for throughput.

Table 1

Feature	Rotor Stator Homogenization	Acoustic Homogenization
Processing Time	5-10 minutes per sample (includes cleaning probe after each sample)	0.5-3 minutes per sample with pulverization, processing time further decreases to <1 minute/sample)
Max Tissue Mass	>3 grams	at least 1 gram
Safety	Aerosol risk, especially of concern with infectious samples	Closed system prevents aerosol exposure
Consistency	Some art to the method—difficult to standardize	Computer controlled energy allows for consistent and controllable delivery for every sample
Scalability	Scaling the system provides no efficiency gains	Easily interfaces with automated systems for improved processing efficiency
Cross-contamination	Carryover from probe tip can contaminate subsequent tissues (especially in cases where high RNA yield tissues such as liver are followed by low yield tissues such as adipose)	Closed system prevents cross-contamination

Methods

Rat and Mouse tissues were dissected and flash frozen in liquid nitrogen and tissues were stored at -80°C until ready for homogenization. Whole livers, muscle tissue (quadriceps), and whole brains were frozen fractured and pieces were weighed prior to homogenization. Tissues were transferred into pre-chilled homogenization tubes, homogenization buffer was added and tissues were homogenized immediately.

Homogenization and Extraction
Polytron homogenization was performed with 10 volumes of Trizol (Invitrogen) added per 100mg tissue (a minimum volume of 2mL of Trizol was used) in 15mL or 50mL screw cap polypropylene centrifuge tubes (Corning). The homogenization was performed as follows: 10,000 rpm for 60 seconds, 25,000 rpm for 60 seconds (max speed), and 10,000 rpm for 30 seconds. After each homogenization the probe was cleaned by rinsing in RNase free water at 25,000 rpm for ~20 seconds, 70% ethanol at 25,000 rpm and again with RNase free water 25,000 rpm. Probe cleaning was essential to minimize the chance of sample cross contamination. The homogenates were stored at -80°C until ready for chloroform extraction.
Covaris homogenization (**Figure 1**) was performed by adding a stabilization buffer to the frozen tissue and then immediately performing the acoustic homogenization for 30-60 seconds using the Covaris E200 focused acoustic device to deliver the appropriate acoustic dose. Samples were stored at -80°C until ready for chloroform extraction.

Sample homogenates from both the Polytron and Covaris homogenizations were then processed through a chloroform extraction and the Total RNA was DNase treated and isolated using silica binding columns. Total RNA was quantified using A260 values obtained with UV spectrophotometry and was assessed for quality by the 28S/18S rRNA ratios obtained on the Agilent (Palo Alto, CA) BioAnalyzer utilizing their Nano Chip Total RNA assay.

Hybridization
Total RNA amplification, labeling and microarray hybridizations were performed as described previously (Hughes et al, Fare et al). Briefly, Agilent ink jet synthesized 60mer arrays containing approximately 25,000 gene spots were used. Samples were set up as fluor reverse pairs (FRP) and hybridizations were performed in custom cartridges for 48 hours followed by washing in 6x SSPE, 0.005% sarkosyl followed by washing in 0.06x SSPE. Slides were dried and then scanned on the Agilent microarray scanner (model G2565AA). Image processing, feature extraction and quantitation were done as described (Marton et al).
Signature compare plots were generated by using the MatLab® software suite (The MathWorks, Inc, Natick, MA) and Rosetta Resolver® gene expression analysis software (Rosetta Biosoftware, Seattle, WA).

Figure 1

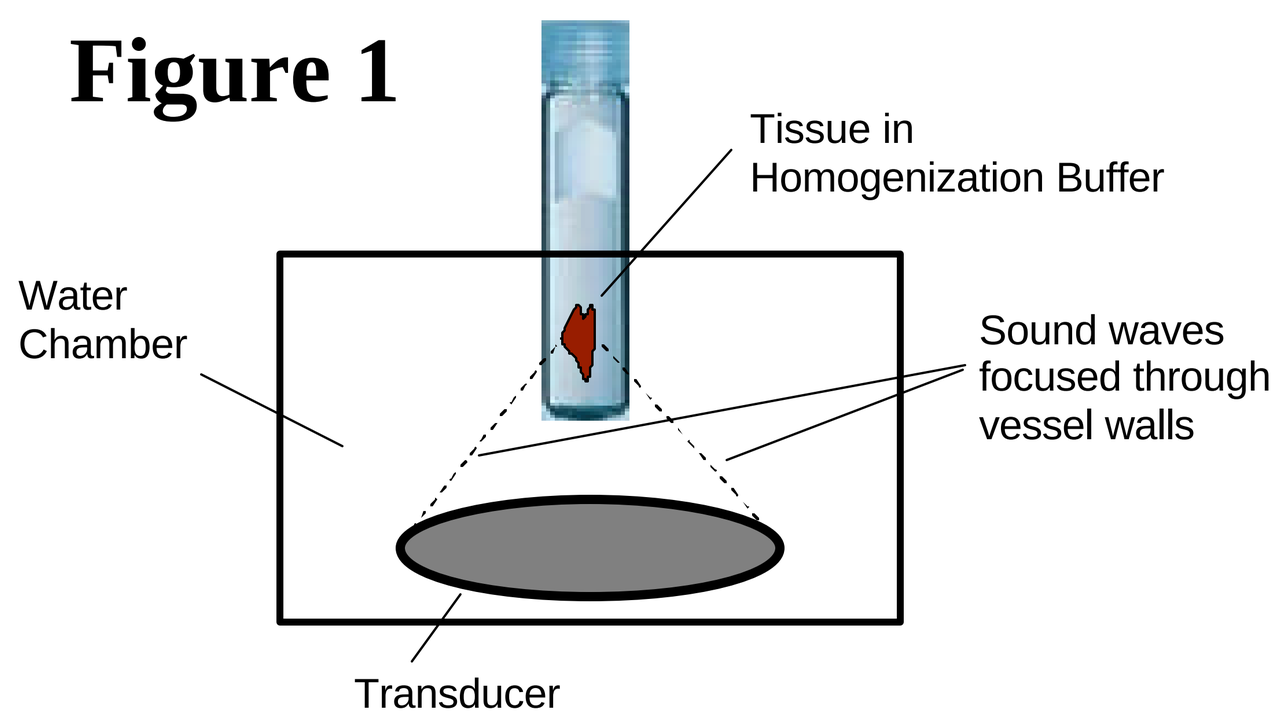
Tissue in
Homogenization Buffer
Water
Chamber
Sound waves
focused through
vessel walls
Transducer

Figure 1 Covaris E200 homogenization chamber. The frozen sample is placed into a chilled homogenization tube, followed by the addition of homogenization buffer and capping of the tube. The tube containing the tissue and buffer is placed into the Covaris E200 water bath and homogenization is initiated using the computer controlled energy delivery system. The transducer directs focused acoustic energy through the walls of the homogenization tube and cavitation events begin to disrupt the tissue, eventually leading to the complete homogenization of the sample. Depending upon the sample type and size, this process is typically completed in less than 60 seconds.

Figure 2 RNA Quality From Rotor Stator and Acoustic Tissue Homogenization

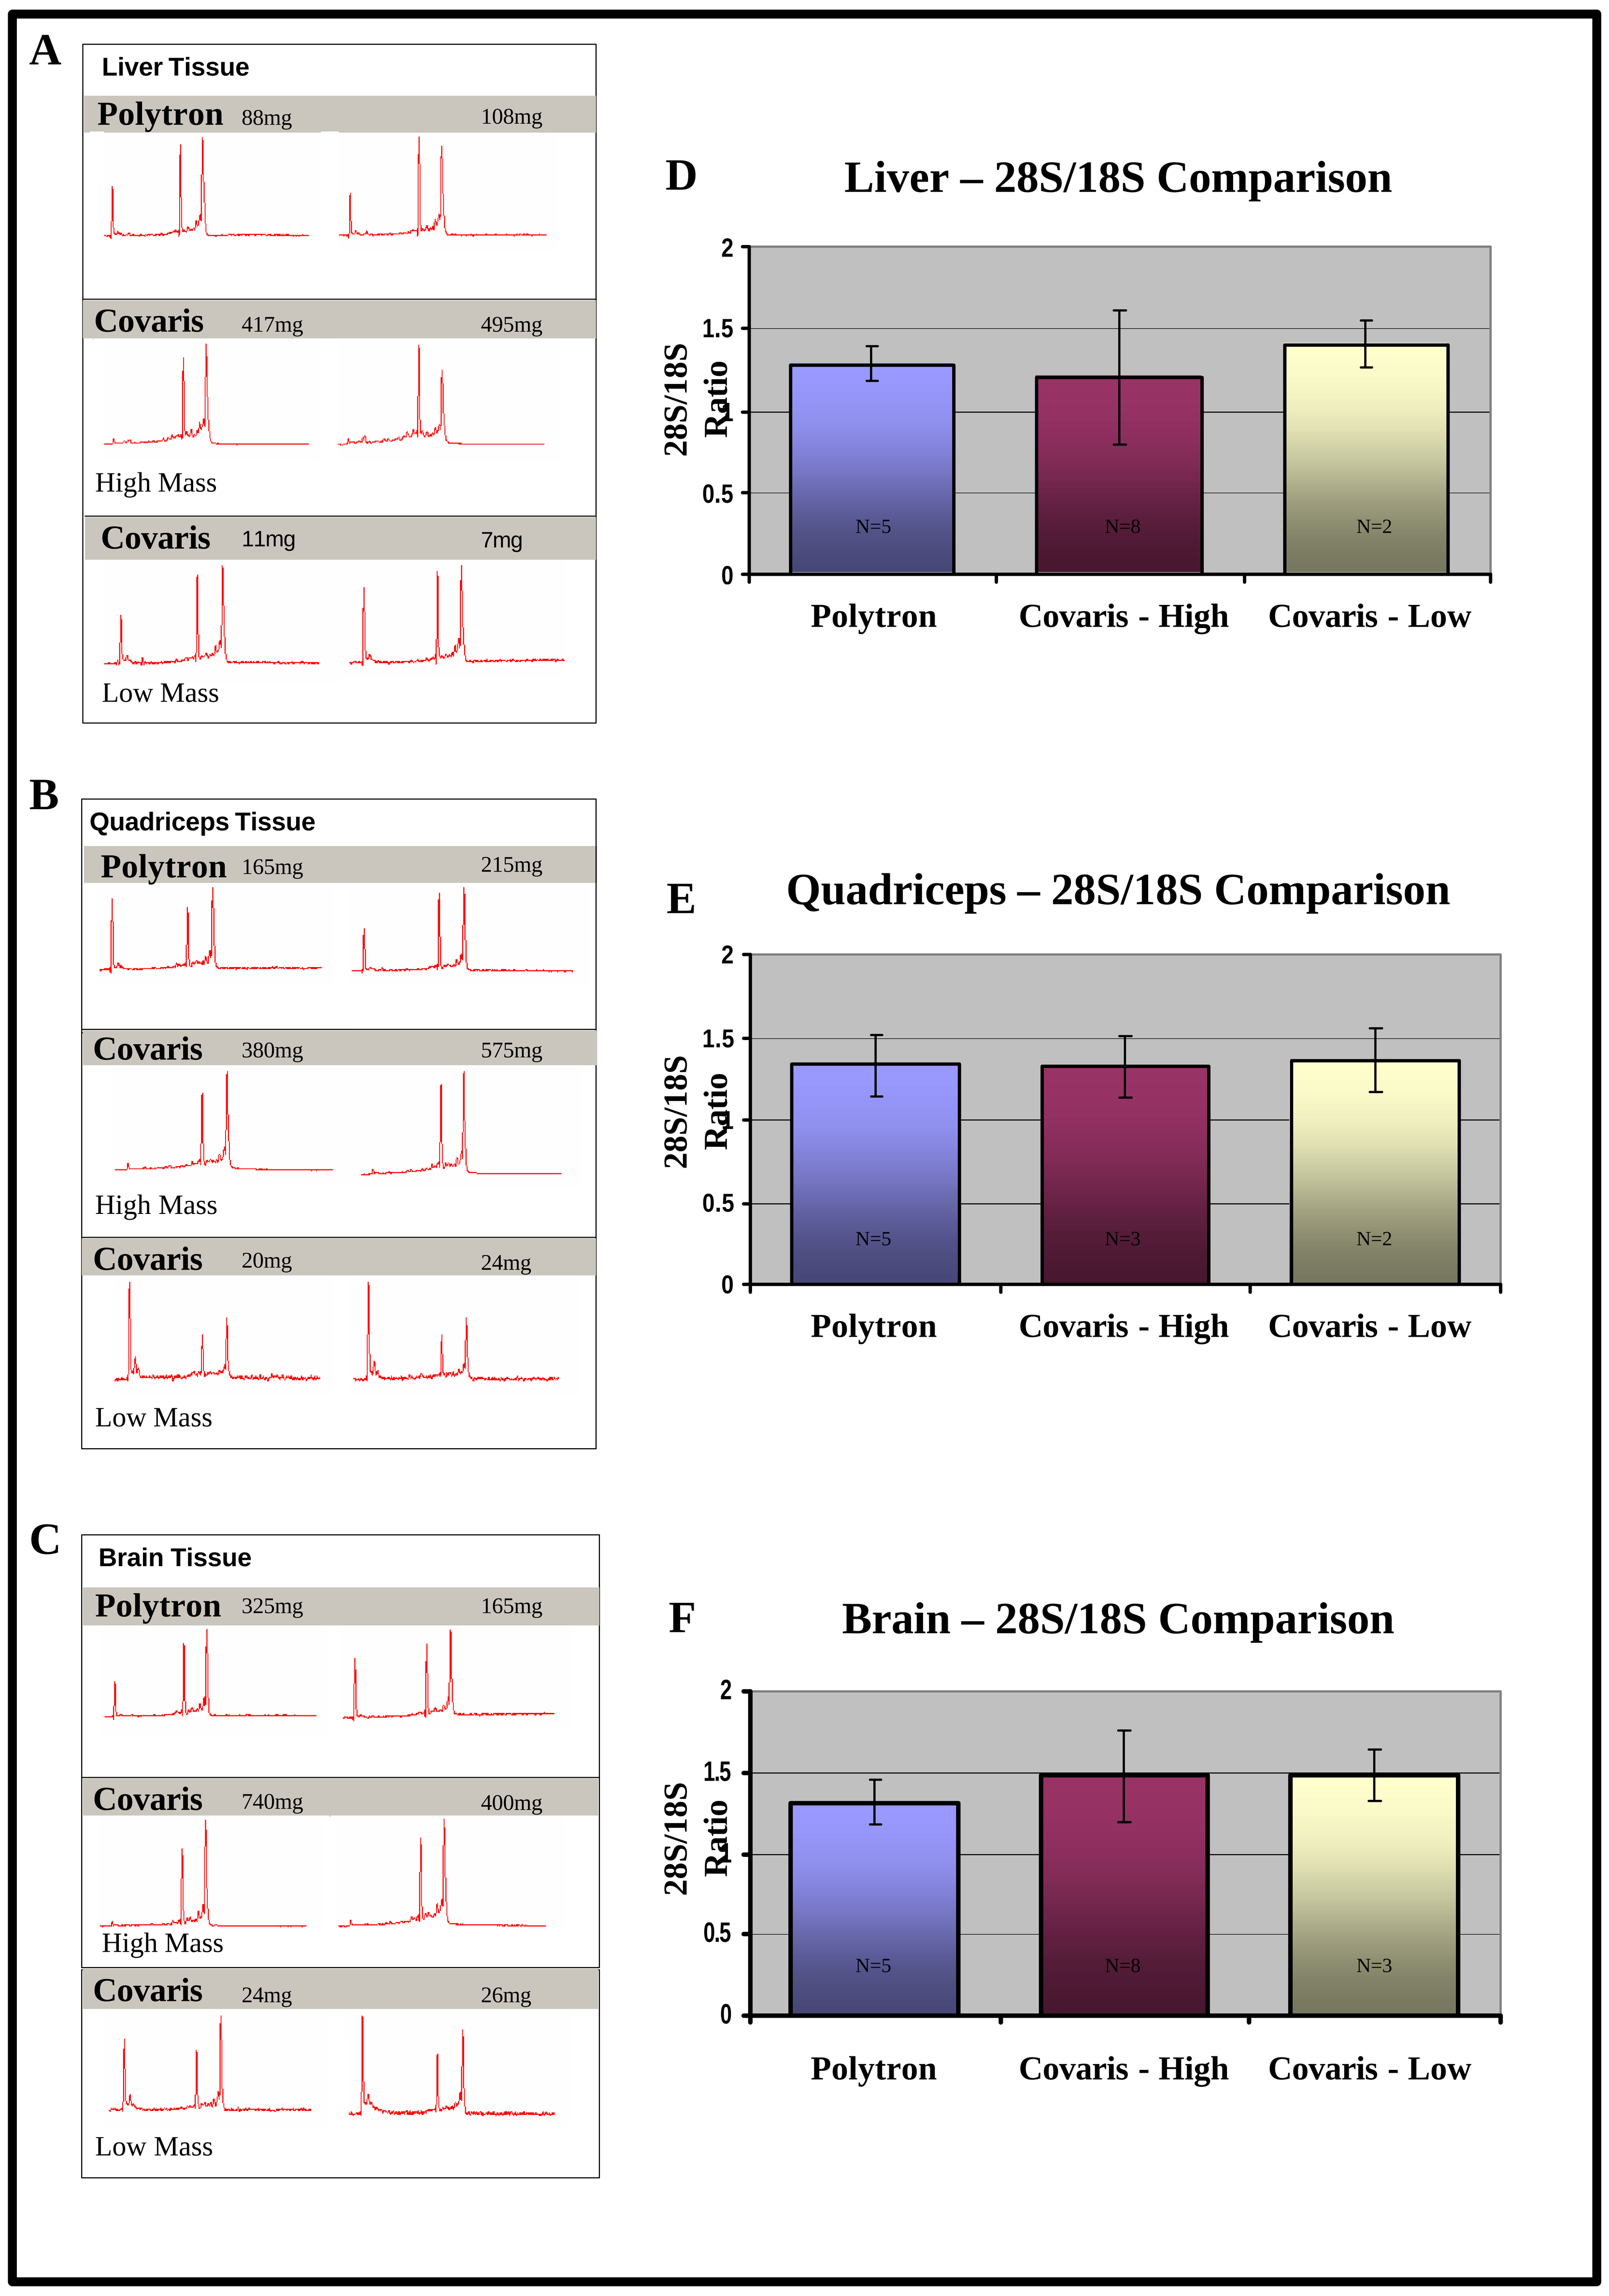
A
Liver Tissue
Polytron 88mg 108mg
Covaris 417mg 495mg
High Mass
Covaris 11mg 7mg
Low Mass
B
Quadriceps Tissue
Polytron 165mg 215mg
Covaris 380mg 575mg
High Mass
Covaris 20mg 24mg
Low Mass
C
Brain Tissue
Polytron 325mg 165mg
Covaris 740mg 400mg
High Mass
Covaris 24mg 26mg
Low Mass
D
Liver – 28S/18S Comparison
28S/18S Ratio
N=5 N=8 N=2
Polytron Covaris - High Covaris - Low
E
Quadriceps – 28S/18S Comparison
28S/18S Ratio
N=5 N=3 N=2
Polytron Covaris - High Covaris - Low
F
Brain – 28S/18S Comparison
28S/18S Ratio
N=5 N=8 N=3
Polytron Covaris - High Covaris - Low

Figure 2 Total RNA Quality Assessment. Panels A-C show Agilent Bioanalyzer traces for the liver, quadriceps, and brain tissue respectively. Panels D-F show the 28S/18S ribosomal RNA peak ratios. The data show there is little or no detectable degradation of RNA. There are acceptable and comparable levels of rRNA ratios in the Polytron and the Covaris E200 homogenized samples.

Figure 3 Reproducibility of Homogenization

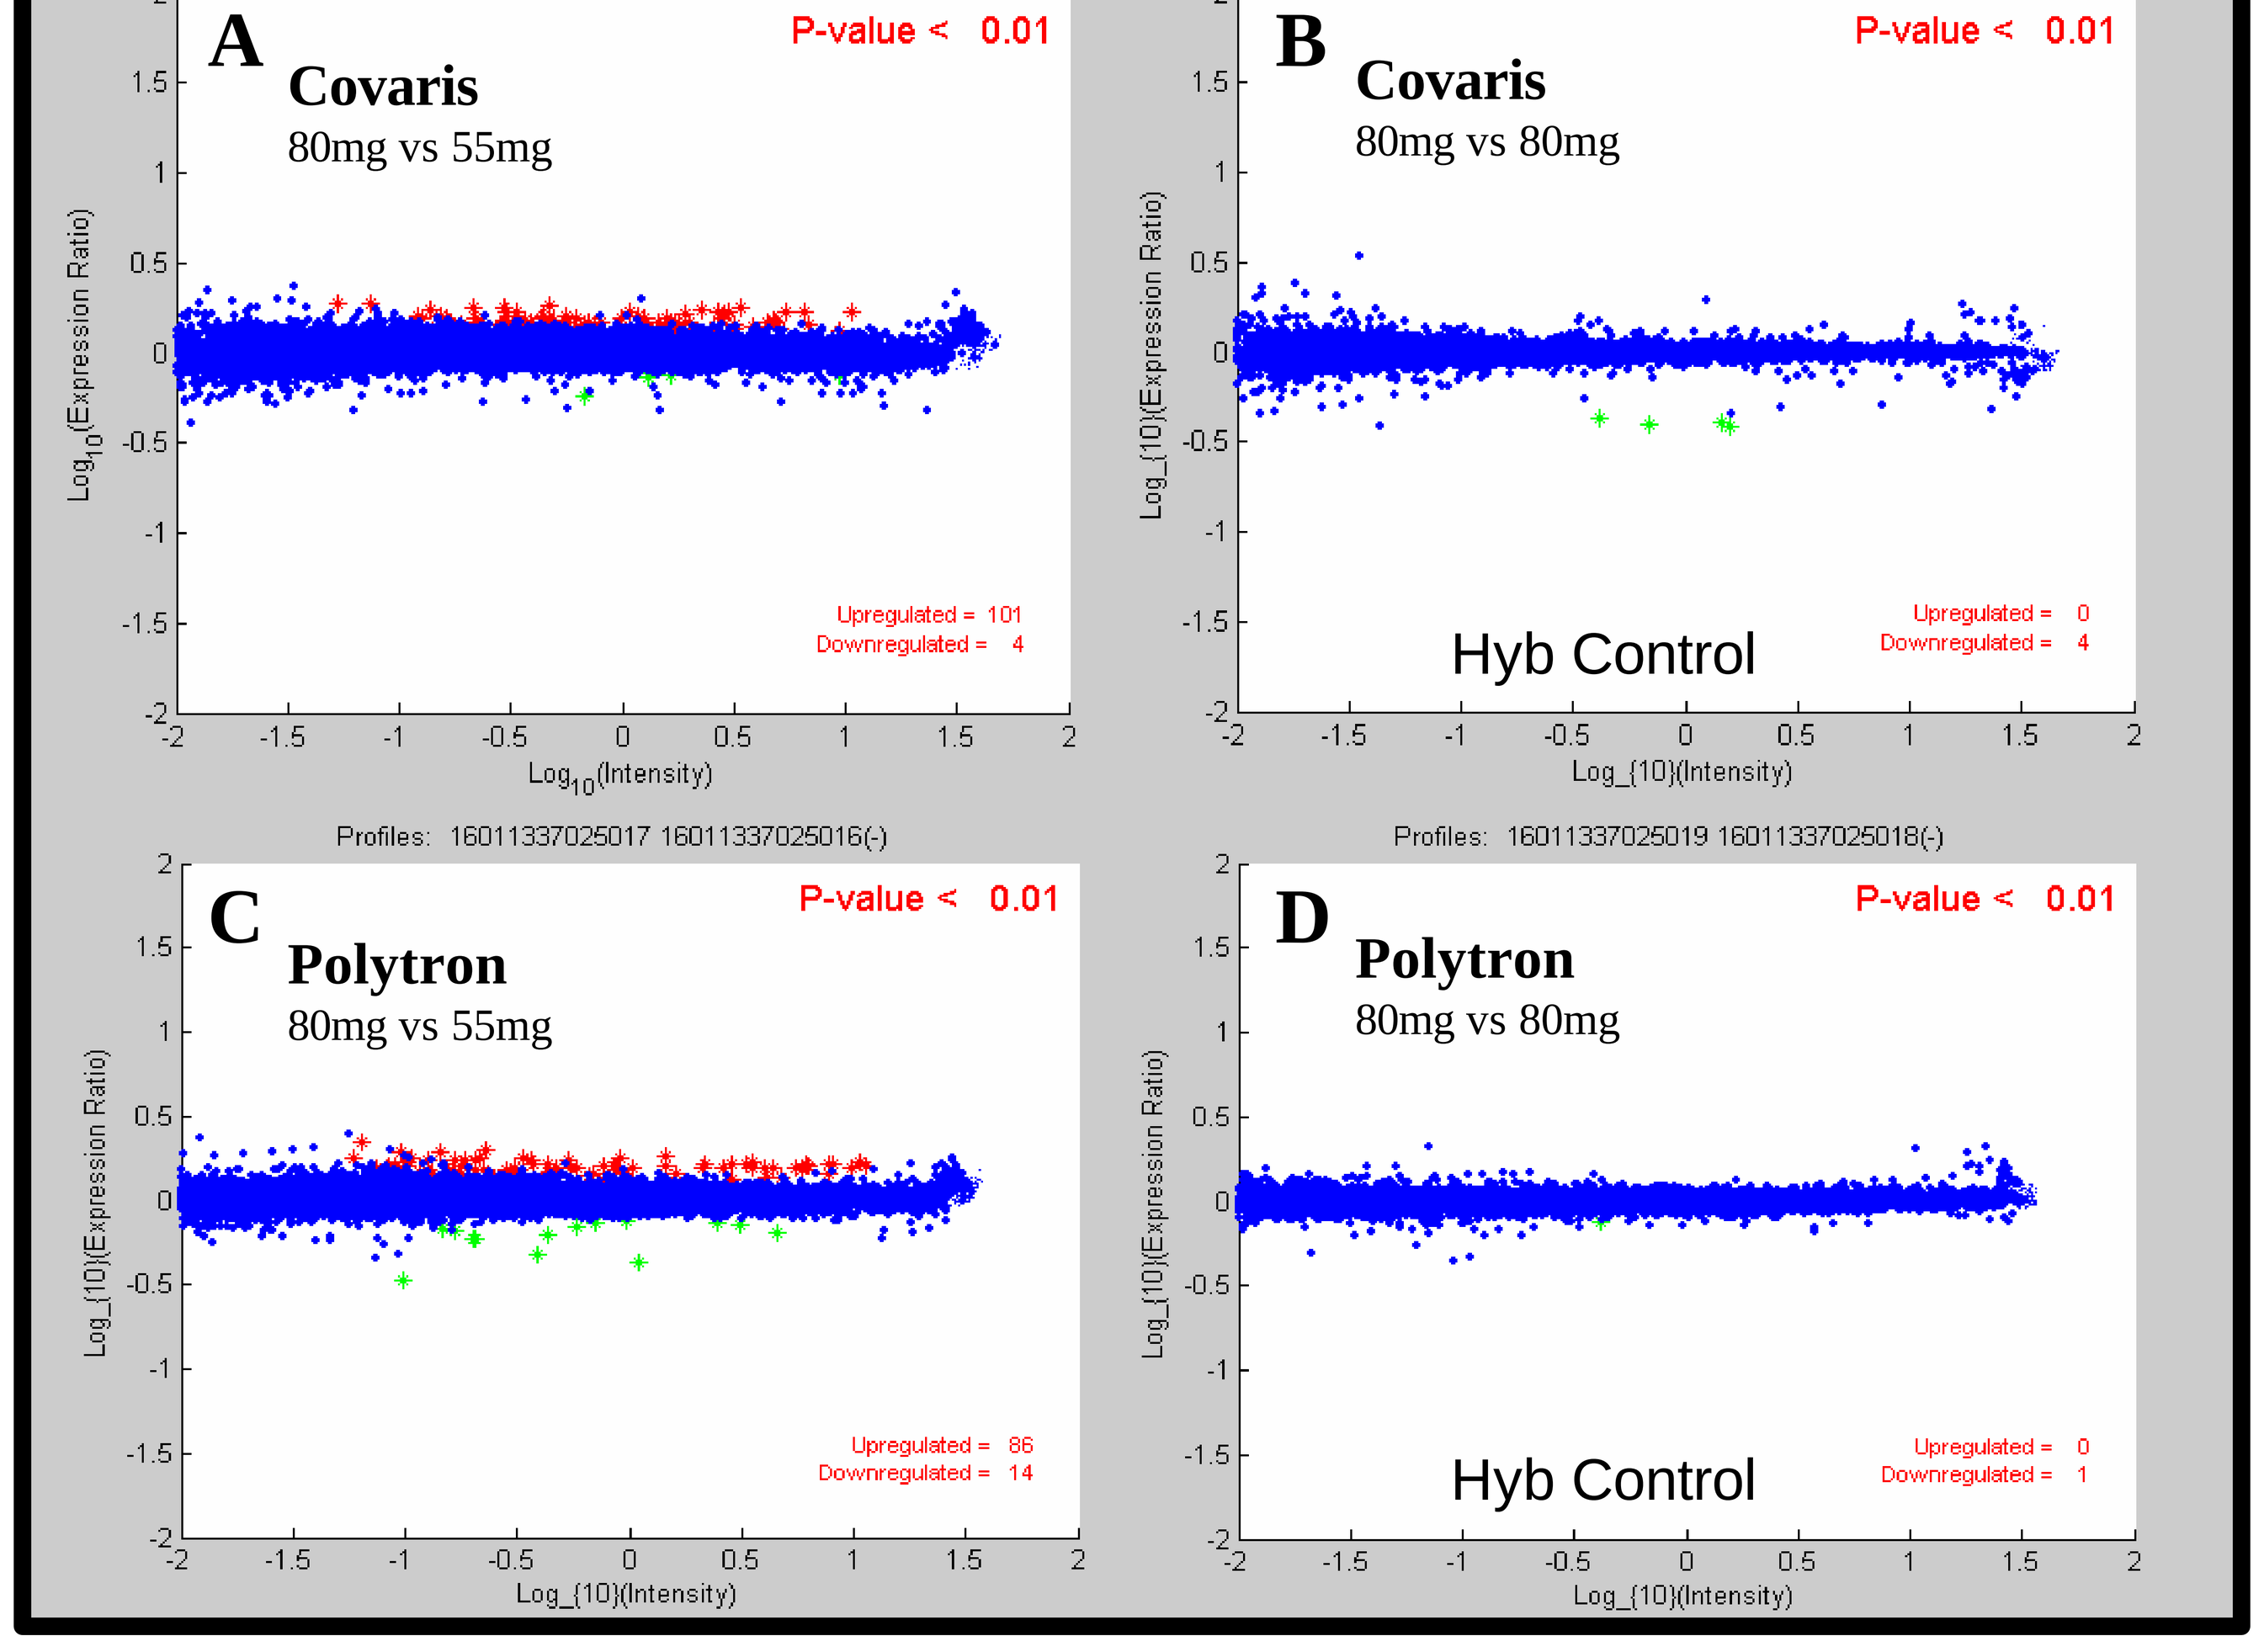
A
Covaris
80mg vs 55mg
P-value < 0.01
Upregulated = 101
Downregulated = 4
B
Covaris
80mg vs 80mg
P-value < 0.01
Hyb Control
Upregulated = 0
Downregulated = 4
C
Polytron
80mg vs 55mg
P-value < 0.01
Upregulated = 86
Downregulated = 14
D
Polytron
80mg vs 80mg
P-value < 0.01
Hyb Control
Upregulated = 0
Downregulated = 1

Figure 3 Reproducibility Demonstrated in Microarray Data. Agilent ink jet synthesized microarrays with ~25,000 60mer gene spots. Red and green spots indicate significant regulations (P-value <0.01). Panels A and B are FRP signature plots for Covaris E200 homogenized samples. Panels C and D are FRP signature plots for Polytron homogenized samples. Panels B and D are amplification and hybridization controls where the same homogenized sample is present in each fluor channel on the individual microarrays making up the FRP. Panels A and C show reproducibility between replicate homogenizations within a homogenization method.

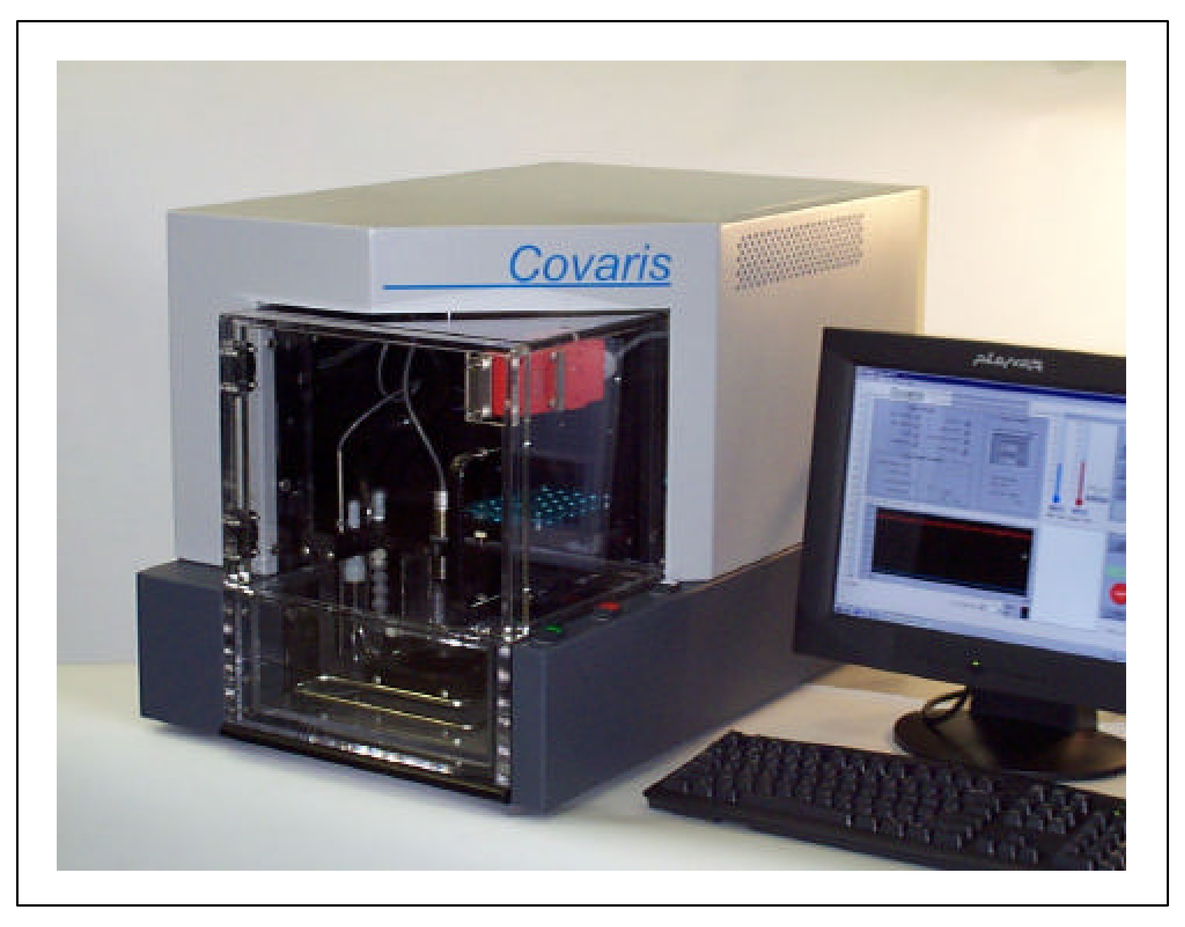
Covaris

Covaris - E200 Focused Acoustic Tissue Disruption System

Results

Total RNA yields were comparable between the Polytron and the Covaris acoustic homogenization method (Table 2). The yields obtained in the comparison were similar to or above expected yield averages for the specific tissues. Expected yield (based upon historical averages or range); Brain 0.15 ug/mg, Muscle 0.4 ug/mg, and Liver average range is 1-3 ug/mg tissue extracted.

Table 2

Tissue	Homogenization Method	Mean Yield ug/mg Tissue	STDEV	N
Brain	Polytron	0.31	0.03	5
Brain	Covaris – High Mass Range	0.35	0.13	4
Brain	Covaris – Low Mass Range	0.83	0.06	3
Quad	Polytron	0.33	0.05	5
Quad	Covaris – High Mass Range	0.24	0.09	3
Quad	Covaris – Low Mass Range	0.42	0.02	2
Liver	Polytron	1.38	0.14	2
Liver	Covaris – High Mass Range	1.45	0.34	8
Liver	Covaris – Low Mass Range	2.48	0.94	2

Total RNA quality was examined using the Agilent Bioanalyzer (BA) (Figure 2). Different ranges of tissue masses (high and low mass ranges) for each tissue were homogenized with the Covaris E200 acoustic system to determine the working range of the instrument (to determine if the RNA obtained from the larger masses would be of equal quality to that obtained from smaller masses). Figure 2 shows that the overall quality of the total RNA obtained from the Covaris E200 homogenizations was comparable to the quality obtained from the Polytron for both the high mass tissue range and the low mass tissue mass range. Additionally, the BA traces were consistent between the higher and lower mass tissue ranges from the total RNA obtained using the Covaris E200.

To further investigate the quality of the total RNA we compared the mean 28S/18S ratios of each homogenization (Figure 2 D-2F). The ribosomal RNA ratios indicate that the Covaris E200 had similar quality for both high and low mass ranges and that each Covaris E200 homogenized tissue mass group had ratios that were comparable to the ribosomal RNA ratios obtained from the Polytron control.

To evaluate the reproducibility of the homogenization we utilized microarrays, which are a very sensitive way to discriminate differences within a homogenization method. The total RNA was amplified and Cy labled in microarray experiments using Fluor-Reversed Pair (FRP) ratio-based approach (Figure 3). Rat Liver RNA from 2 separate homogenizations was compared for each homogenization method. The tissue from the same homogenization within each method was hybridized as a control FRP where little or no regulations should be present. Comparisons were then made to the separate homogenization FRP noise (as represented by total significant regulations). The hybridization results indicate that the homogenization methods contribute similar noise on a microarray.

Discussion

This study has explored the use of non-contact, focused acoustic energy as a method for tissue homogenization. Homogenates from this procedure were then extracted and input into high throughput total RNA isolation utilizing silica binding plates. The quality of total RNA for both high mass (up to 740 mg of rat brain) and low mass samples (as low as 7 mg of rat liver) obtained with the Covaris E200 homogenization, were comparable to the quality obtained from the same sample types, and in some cases fragments from the exact same tissue, as using the Polytron technique.

The yields obtained by the acoustic tissue disruption process were also within expected parameters for the tissue types evaluated. Of particular interest is the fact that in theory the yields from acoustically homogenized tissue could be higher because the tissue is homogenized to a much finer particulate than that of the Polytron. The larger number of fine tissue fragments, with the acoustic homogenization, would cause increased surface area and should allow for a more rapid and effective RNA preservation and extraction (e.g. guanidinium/phenol). In addition, the finer particulate size of acoustically homogenized tissue should prevent clogging of the silica binding matrix that is used to isolate the total RNA. The acoustic energy may also shear genomic DNA to an extent which would alleviate the need for DNase treatments in the RNA isolation, although in this study we included a standard DNase treatment in the isolation of RNA from all samples.

The reproducibility of the homogenization methods, were evaluated by analyzing microarray data using RNA obtained from replicate rat liver homogenates prepared by the Covaris E200 or Polytron. "Same vs Same" hybridizations using a FRP ratio-based approach were set up to look at the number of false gene regulations or noise, when RNA isolated from two independent homogenizations within a method were used. The data demonstrated that there was similar noise introduced by comparing different homogenizations within each method. The low noise values represented by the total number of significant (p <0.01) gene regulations supports that both methods are very reproducible.

In conclusion, using the Covaris acoustic tissue disruption process we have demonstrated that we were able to obtain comparable yield and quality of isolated total RNA compared to the Polytron method for brain, liver, and quadriceps mammalian tissue. Evaluation of microarray gene expression data from RNA isolated from replicate homogenizations from the same rat liver have demonstrate that the Covaris E200 is comparable to Polytron replicate homogenizations with respect to reproducibility. Therefore, based upon these evaluations, the Covaris focused acoustic process is a feasible and attractive alternative to the rotor stator method for tissue homogenization, where the extracted nucleic acid products will be used for sensitive downstream applications such as microarray analysis.

References

Fare, T.L. ; Coffey, E.M.; Dai, H.; He, Y.D.; Kessler, D. A.; Kilian, K.A.; Koch, J.E.; LeProust, E.; Marton, M.J.; Meyer, M.R.; Stouhton, R.B., Tokiwa, G.Y.; Wang, Y. (2003) Effects of Atmospheric Ozone on Microarray Data Quality *Anal. Chem*., **75**, 4672-4675

Hughes, T.R.; Mao, M; Jones, A.R.; Burchard, J.; Marton, M.J.; Shannon, K.W.; Lefkowitz, S.M.; Ziman, M.; Schelter, J.M.; Meyer, M.R.; Kobayashi, S.; Davis, C.; Dai, H.; He, Y.D.; Stephaniants, S.B.; Cavet, G.; Walker, W.L.; West, A.; Coffey, E.; Shoemaker, D.D.; Stoughton, R.; Blanchard, A.P.; Friend, S.H.; Linsley, P.S. (2001)Expression profiling using microarrays fabricated by an ink-jet oligonucleotide synthesizer. *Nat Biotechnol*;**19**:342-347

Marton, M.J.; DeRisi, J.L.; Bennett, H.A; Iyer, V.R.; Meyer, M.R.; Roberts, C.J.; Stoughton, R; Burchard, J.; Slade, D.; Dai, H.; Basset, D. E. Jr.; Hartwell, L.H.; Brown, P.O.; Friend, S.H. (1998) Drug target validation and identification of secondary drug target effects using DNA microarrays *Nat. Med.*, **4** (11), pp 1293 –1301.